Proteomics and Transgenic Animal Models of Alzheimer's Disease

E. McGregor*[1], V.Patel [1], J.A. Westbrook[1], N.Leeds[1], H.Byers[1], J.Campbell[1], K.Leung[1], C.Fitzer-Attas[2] & M.Ward[1].
[1]Proteome Sciences plc, PO45, Institute of Psychiatry, Kings College, London, UK.
[2]Mindset BioPharmaceuticals, Jerusalem, Israel.

INTRODUCTION

Alzheimer's disease (AD), the most common cause of dementia in older individuals, is a debilitating neurodegenerative disease for which there is no cure. Misappropriate processing of the amyloid precursor protein (APP), or failure to clear the amyloid beta (Aβ) peptide fragments, results in toxic elevations of this peptide which are deposited throughout the brain as insoluble fibrillar aggregates. Resulting plaques are particularly concentrated in the hippocampus. Identification of AD causing mutations in APP, presenilin (PS) 1, and PS2 that affect Aβ levels further supports the critical role for Aβ in the development of AD. Ongoing elucidation of the mechanisms that underlie the pathogenesis of AD will facilitate the identification and development of disease-modifying drugs and effective sensitive methods for early detection and diagnosis of, or predisposition to, AD.

AIMS

- Identify new diagnostic markers of AD
- Identify new therapeutic targets of AD

TRANSGENIC MOUSE MODEL

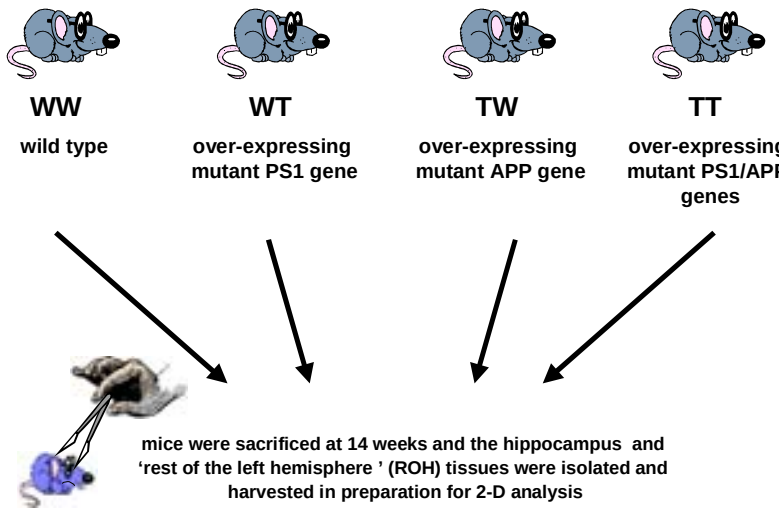
WW
wild type
WT
over-expressing mutant PS1 gene
TW
over-expressing mutant APP gene
TT
over-expressing mutant PS1/APP genes
mice were sacrificed at 14 weeks and the hippocampus and 'rest of the left hemisphere ' (ROH) tissues were isolated and harvested in preparation for 2-D analysis

Figure 1. Transgenic mouse model of AD
We have used single (over-expressing mutant alleles of APP or PS1) and double (APP/PS1) transgenic mouse models to investigate the pathogenesis of AD. APP mice exhibit age-dependent accumulation of brain Aβ deposits. PS1 mice show increased production of the Aβ1-42 peptide, but do not form amyloid deposits upon aging, presumably because the levels of Aβ do not reach the level required to start the aggregation process. Double transgenic mice (APP/PS1) have many features consistent with the pathology of human AD, e.g. dense plaque formation, hyperphosphorylated tau, and importantly, severe cognitive and behavioural impairment.

SAMPLE PREPARATION

Samples were prepared according to Weekes *et al* [1] and Heinke *et al* [2]. Hippocampus and Rest of left hemisphere (ROH) tissues were solubilised in 9.5M (w/v) urea, 2% (w/v) CHAPS, 1% DTT (w/v), 0.8% (v/v) Pharmalyte, pH 3-10, 150 μl ml^{-1} Mini Complete Protease Inhibitor Cocktail tablets (Roche Molecular Biochemicals). Solubilised protein was harvested and the protein concentration determined using a modification of the method described by Bradford [3].

PROTEIN SEPARATION: 2-D PAGE

2-D PAGE was performed according to Weekes *et al* [1] and Heinke *et al* [2]. The 2-D system comprised a linear pH 4-7 and pH 6-9 in the first dimension (180 mm immobilised pH gradient strips (IPG strips), Amersham Biosciences) for hippocampus and ROH with 12%T SDS polyacrylamide gel electrophoresis in the second dimension (Figure 2). For all pH 6-9L gradients, strips were rehydrated in DeStreak rehydration solution (Amersham Biosciences) and samples cuploaded at the anode. Analytical gels, run for subsequent quantitative computer image analysis, were loaded with 100 μg of protein.

Micro-preparative gels were run for both sample types using both pH gradients with total protein loadings of 400 μg. Micro-preparative total protein spot profiles were visualised using a modified PlusOne silver stain (Amersham Biosciences) [4].

All silver stained gels were scanned at 100 μm resolution using a Molecular Dynamics Personal SI Laser Densitometer (Sunnyvale, CA, USA).

2-D

	hippocampus		ROH	
	pH 4-7L *n*	pH 6-9L *n*	pH 4-7L *n*	pH 6-9L *n*
WW	5	5	10	10
WT	5	5	9	9
TW	3	4	8	8
TT	4	4	10	10

Figure 2. 2-D analysis of hippocampus and ROH

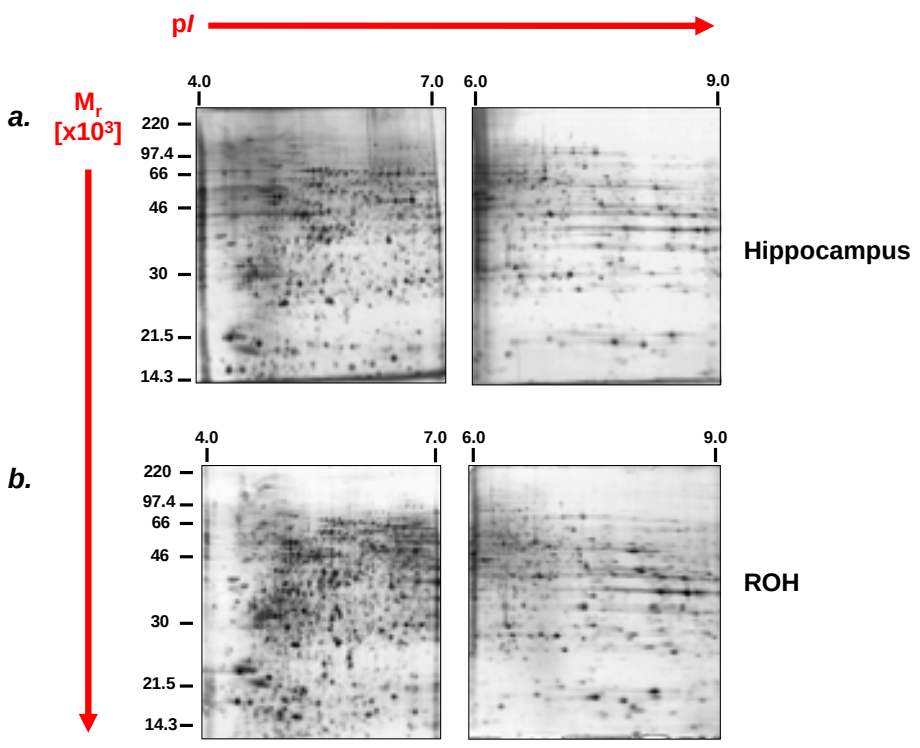
pI
a.
Mr [x10³]
4.0
7.0
6.0
9.0
220
97.4
66
46
30
21.5
14.3
Hippocampus
b.
ROH

Figure 3. 2-D gel protein profiles
Example 2-D gel images for pH 4-7L and 6-9L gradients of ***a.)*** hippocampus and ***b.)*** ROH tissue

2-D GEL IMAGE ANALYSIS

Scanned silver stained gel images were analysed quantitatively using the specialised software Progenesis™ Workstation, version 2003.02 (Nonlinear Dynamics Ltd). Briefly, images were automatically spot detected, warped and matched. All images underwent manual editing of detected spots and manual matching to optimise these two parameters. Following background subtraction and normalisation of all protein spot volumes, protein spot data was exported to Excel for statistical analysis.

Individual two-group comparisons, using the Student *t*-test ($p \leq 0.05$), were performed, comparing each transgenic group against wildtype: WW vs WT; WW vs TW; WW vs TT (Table 1). The spot criteria used was that a matched spot had to be present in at least 60% of gels of a given transgenic group

STUDENT *t*-TEST RESULTS

Comparison	Spot No.	Fold change	p ≤ 0.05
Hippocampus, pH 4-7L			
WW vs TT	240	2.13 ↑	0.01
	458	11.0 ↑	0.01
	542	2.06 ↓	0.01
	546	2.01 ↓	0.04
	669	2.65 ↓	0.03
	756	2.32 ↓	0.01
	763	3.88 ↓	0.03
	903	2.36 ↓	0.01
	908	2.14 ↓	0.002
	1021	3.25 ↓	0.02
	1052	2.19 ↓	0.04
	1056	2.24 ↓	0.01
	1189	2.05 ↓	0.01
	1199	2.50 ↓	0.02
	1224	2.25 ↓	0.02
	1401	3.27 ↓	0.003
	1514	2.56 ↓	0.05
	1518	2.99 ↓	0.05
	1571	2.96 ↓	0.03
	1653	2.13 ↓	0.009
	1683	2.95 ↓	0.04
	1848	2.77 ↓	0.008
Hippocampus, pH 4-7L Common spots between groups			
WW vs TT	1976	3.5 ↓	0.003
WW vs TW		2.0 ↓	0.004
WW vs TT	614	2.1 ↑	0.004
WW vs WT		1.8 ↑	0.04
WW vs TW	1341	3.2 ↓	0.003
WW vs WT		2.0 ↓	0.01
WW vs TW	1554	2.3 ↑	0.01
WW vs WT		1.7 ↑	0.03
WW vs TW	1498	1.3 ↓	0.03
WW vs WT		1.1 ↓	0.007
WW vs TT	1585	2.3 ↓	0.02
WW vs TW		2.6 ↓	0.02
Hippocampus, pH 6-9L			
WW vs TW	971	2.1 ↑	0.01
	1191	2.4 ↑	0.01
	1303	1.9 ↑	0.04
Hippocampus, pH 6-9L Common spots between groups			
WW vs TW	884	1.7 ↓	0.01
WW vs WT		1.9 ↓	0.02
WW vs TW	980	1.4 ↓	0.01
WW vs WT		1.4 ↓	0.02
WW vs TW	1017	2.5 ↓	0.03
WW vs WT		2.6 ↓	0.02
WW vs TW	1023	1.2 ↓	0.04
WW vs WT		1.3 ↓	0.03
WW vs TT		1.3 ↓	0.02
ROH, pH 4-7L Common spots between groups			
WW vs TW	696	1.7 ↑	0.02
WW vs WT		1.7 ↑	0.04
WW vs TW	372	1.4 ↑	0.03
WW vs TT		1.3 ↑	0.03
WW vs WT	1823	1.2 ↓	0.05
WW vs TT		1.3 ↓	0.01
WW vs WT	7427	1.4 ↑	0.03
WW vs TT		1.5 ↑	0.05
WW vs WT	1566	1.5 ↑	0.05
WW vs TW		1.4 ↑	0.02
WW vs TT		1.5 ↑	0.02
ROH, pH 6-9L Common spots between groups			
WW vs TT	1023	1.7 ↑	0.006
WW vs TW		1.9 ↑	0.02
WW vs TT	1661	1.2 ↑	0.02
WW vs TW		1.3 ↑	0.03
WW vs TT	1758	1.3 ↑	0.02
WW vs TW		1.4 ↑	0.03
WW vs TT	1892	1.5 ↑	0.04
WW vs TW		1.8 ↑	0.03

Table 1.
The table above presents all significant changes in protein spot expression found in the Student *t*-test comparisons previously described.

The phrase 'common spots between groups' refers to protein spots which were found to exhibit similar changes in expression in comparisons between the WT, TW and TT transgenic groups and wildtype, WW.

The functions of identified proteins found to significantly change in their expression include and involve, glycolysis, actin filament dynamics, neurotransmitter release, protein folding and oxidative stress.

PRINCIPAL COMPONENT ANALYSIS (PCA)

PCA was applied to all datasets. Analysis was performed initially on all transgenic groups and subsequently on just WW and TT. Spots included as variables were those present in at least 60% of gels from any one of the groups.

Inspection of the majority of the PCA scores plots did not reveal clear clusters correlating with wildtype or transgenic group membership. However, an exception to this was the comparison of WW vs TT, hippocampus, pH 4-7. There was clear separation of these two groups as illustrated in the PCA scores plot (Figure 4).

This visual display of data correlates with the Student *t*-test results for this comparison which revealed the most significant changes in protein spot expression. When we look to see where spots, found to be significant by Student's t-test, fall on the loadings plot, their locations are as expected, i.e at the very top and bottom of the plot, at the extremes of the second principal component (Figure 4).

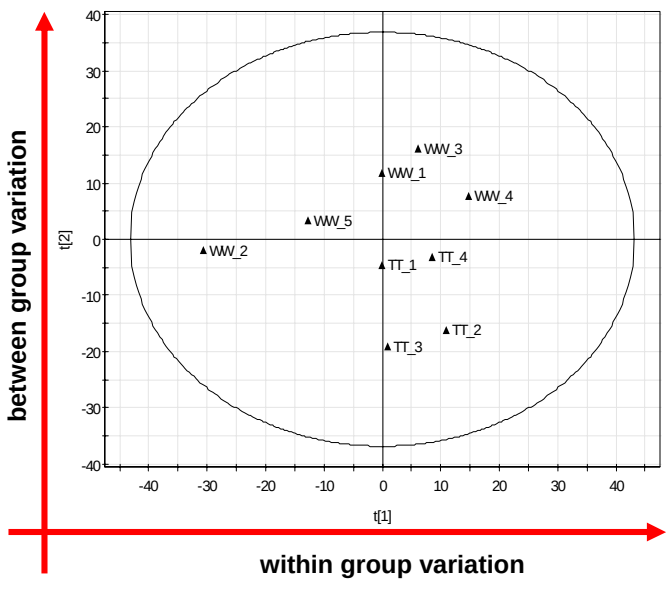
between group variation
within group variation
t[2]
t[1]
40
30
20
10
0
-10
-20
-30
-40
WW_1
WW_2
WW_3
WW_4
WW_5
TT_1
TT_2
TT_3
TT_4

Figure 4.
PCA scores plot showing the first two principal components (within group and between group variation) in the analysis of WW vs TT, hippocampus, pH 4-7.

MS ANALYSIS

After quantitative image analysis spots of interest were excised from silver stained micro-preparative gels for protein identification. In-gel reduction, alkylation and digestion with trypsin were performed prior to subsequent analysis by mass spectrometry [5].

LC/MS/MS is a very powerful technique for the sequencing and identification of peptides as well as having the capabilities of obtaining additional information about post-translational modifications e.g. phosphorylation. Peptides are first desalted, concentrated and resolved by reversed-phase chromatography. This vastly increases the sensitivity of the technique.

Chromatographic separations were performed on a 75 µm, reversed-phase (C18) PepMap column using an Ultimate LC system (Dionex). A gradient of acetonitrile in 0.05% formic acid was delivered to elute the peptides at a flow rate of 200 nl/min. Peptides were ionised by electrospray ionisation using a Z-spray source fitted to a Q-Tof-*micro* (Waters Corp., USA). The instrument was set to run in automated switching mode, selecting precursor ions based on their intensity. The MS/MS analyses were conducted using collision energy profiles that were chosen based on the m/z and the charge state of the peptide. The MS/MS data was processed into peak lists and analysed using the Mascot searching algorithm (Matrix Science).

FUTURE WORK

Future work will include analysis of protein spots, found to exhibit differential expression, by mass spectrometry from the pH 6-9L set of gels for the ROH tissue, which is being undertaken presently.

Serum samples from wildtype (WW) and all transgenic mice (WT, TW, TT) in this study have been collected and are being analysed by SELDI. These results will be compared against differentially expressed proteins identified from whole brain tissue.

REFERENCES

1. Weekes, J. *et al*, *Electrophoresis,* 20 (4-5):898-906, 1999.
2. Heinke, M.Y. *et al*, *Electrophoresis,* 20 (10):2086-2093, 1999.
3. Bradford, M.M. *Anal. Biochem.,* 72, 248-254, 1976.
4. Yan, J.X. *et al*, *Electrophoresis,* 21:3666-3672, 2000.
5. Shevchenko, A. *et al*, *Anal. Biochem.,* 68, 850-858, 1996.

ACKNOWLEDGEMENTS

We would like to thank the Britech Foundation Ltd for providing grant funding to support this project.

britech